Reducing Clinic No-Show Rate
North Central Public Health District
Wasco, Sherman and Gilliam Counties, OR

Public Health
Prevent. Promote. Protect.
NORTH CENTRAL PUBLIC HEALTH DISTRICT
"Caring For Our Communities"

Presenter:
Faith Purvis, RN

The Problem:

North Central Public Health District has had a high no-show rate for reproductive health clinic appointments, leading to wasted resources and reduced effectiveness of services.

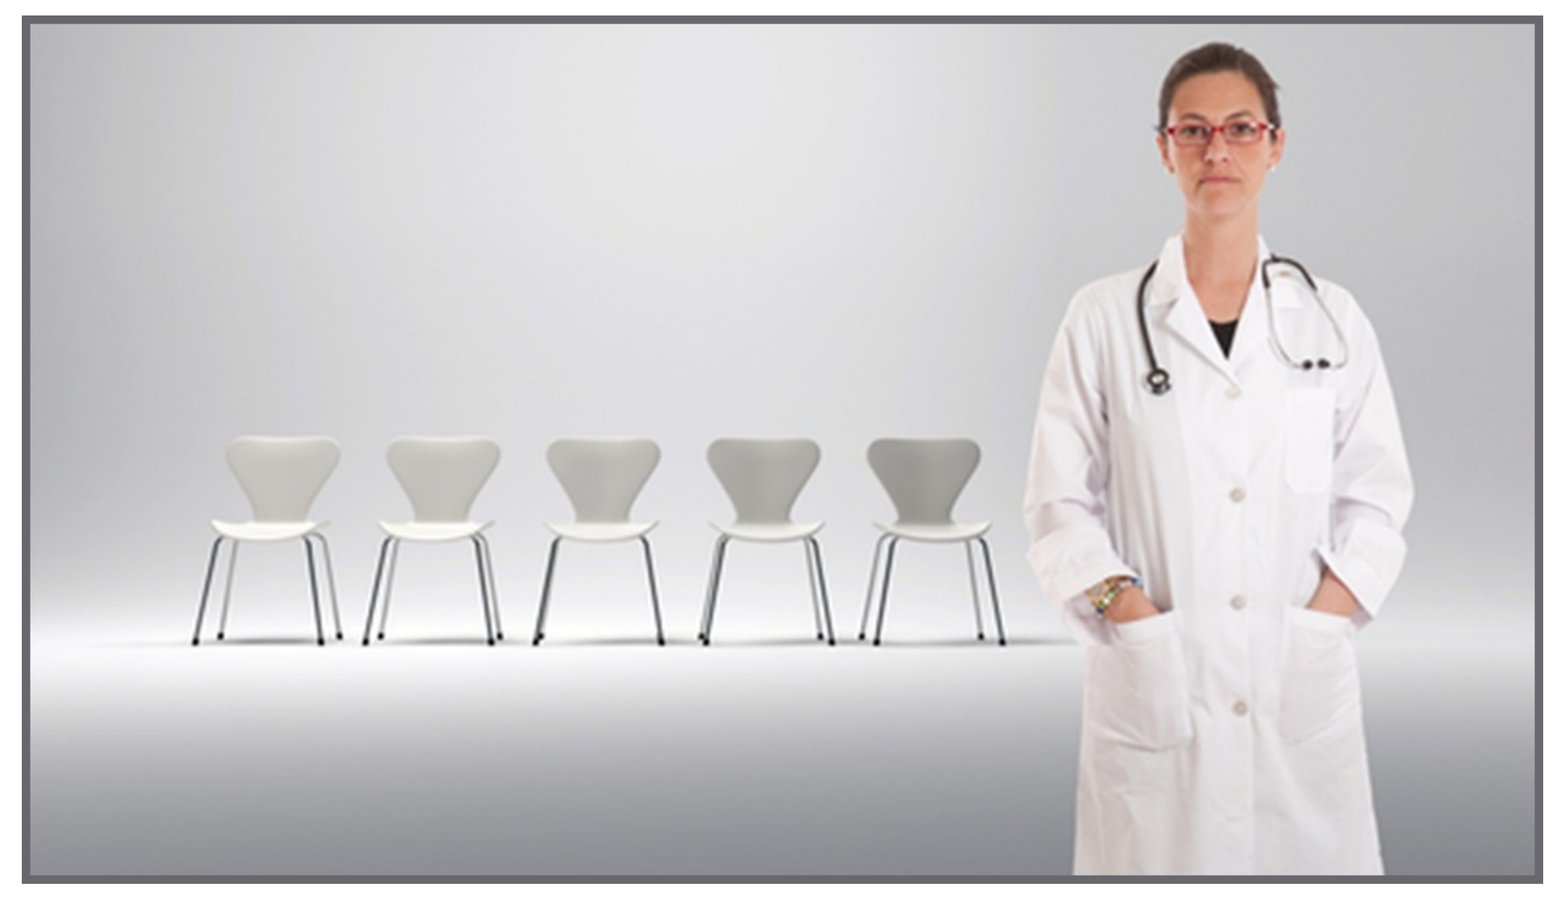

The Goal: Reduce and maintain no-show rate to at or below the state average of 20%.

Phase I:

In April 2012, the QI team convened. We reviewed and then implemented some strategies that had worked for other clinics addressing the same problem. We then monitored the data for 6

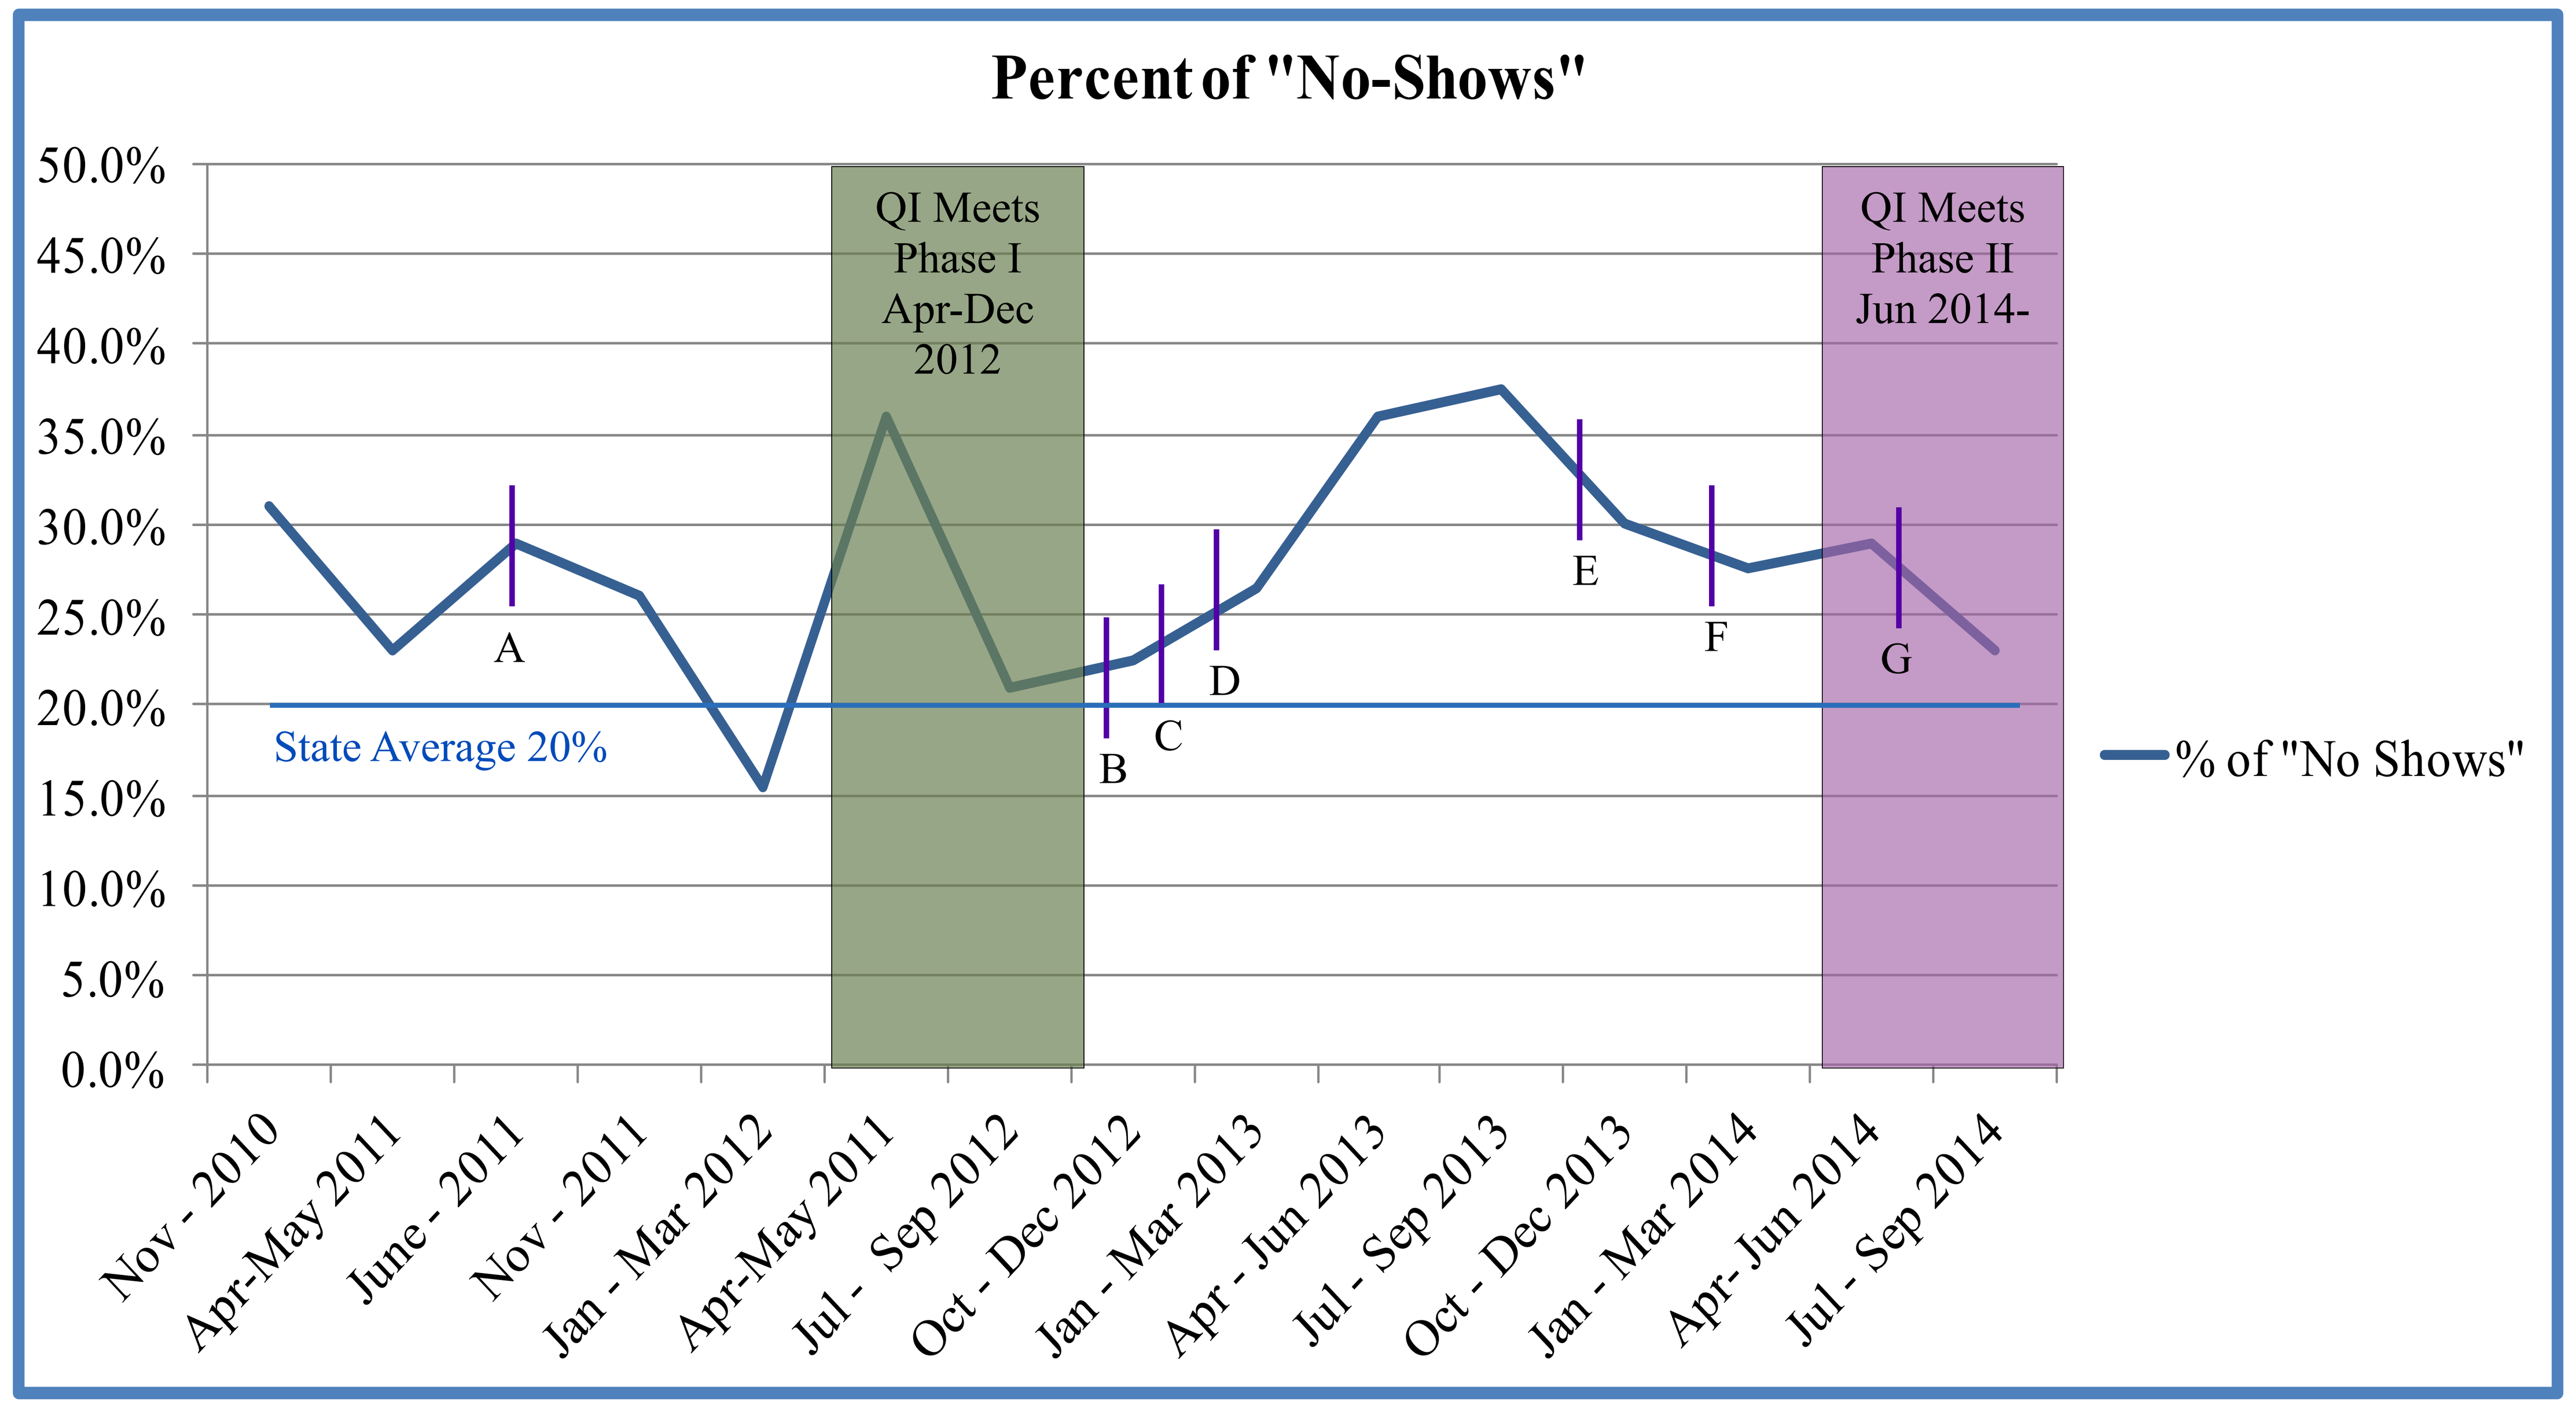
Percent of "No-Shows"
50.0%
45.0%
40.0%
35.0%
30.0%
25.0%
20.0%
15.0%
10.0%
5.0%
0.0%
QI Meets Phase I Apr-Dec 2012
QI Meets Phase II Jun 2014-
State Average 20%
A
B
C
D
E
F
G
% of "No Shows"
Nov - 2010
Apr-May 2011
June - 2011
Nov - 2011
Jan - Mar 2012
Apr-May 2011
Jul - Sep 2012
Oct - Dec 2012
Jan - Mar 2013
Apr - Jun 2013
Jul - Sep 2013
Oct - Dec 2013
Jan - Mar 2014
Apr- Jun 2014
Jul - Sep 2014

A: September 2011: Transition to New Provider
B: Jan 2013: Clinic Days Change from MWF to M/W
C: Jan-Feb 2013: Transition to New Provider
D: Apr 2013: Clinic Days Change from M/W to M/T
E: Oct 2013: Affordable Healthcare Act rollout
F: Jan 2014: Oregonians with Food Stamps automatically enrolled into OHP
G: August 2014: Reminder call timing changed

Phase II:

In 2014, the QI team met again. We asked, what possible confounding factors and changes have occurred?

Internal changes:

- January 2013: Transitioned to a new provider.
- January 2013: Reduced our clinic days from 3 to 2 days/week.
- Because of clinic day change, reminder calls no longer being made two days in advance. Instead calls made Friday for Monday appts, and Monday for Tuesday appts.
- Follow-up calls no longer being made to ask why appt missed.

External changes:

- Many formerly uninsured clients started gaining insurance through the Affordable Healthcare Act (ACA).
- Managed Care/CCO's encouraging more services through clients' designated primary care providers.

Other Factors???

Reducing Clinic No-Show Rate QI Phase 1: April-Dec 2012

Plan	Do	Check	Act
Improve the Reproductive Health Clinic Show Rate	1. Make reminder calls two days prior to appt (previously calls were made one day before, or sometimes not at all). 2. Use language emphasizing importance of keeping appt 3. Clarify what appt will entail, dispel misconceptions. (i.e. "annual exam" does not always require a pelvic exam) 4. Follow-up call to no-show clients to better understand barriers. 5. Assess days and demographics of no-shows. 6. Hold a raffle for clients who are on time for appt.	-Prepare and compare data reports over the six months. -Assess success of raffle: most clients not interested. - Assess success of other strategies: They appear effective. **Findings at end of Dec 2012: Missed appointments reduced from 36% to 20%**	-Continue measures 1-4 -Discontinue measures 5-6 -Observe trends and reassess data as needed.

At end of 2012 we declared success: We lowered no-show rate to 20%.

Time to **Celebrate!**

But wait …. What about the 2nd half of our goal: *maintaining* 20%?

As the graph shows, we only briefly reached our goal before the rate started rising again.

The upshot: **QI truly needs to be an ongoing process!**

The QI team decided we needed to re-convene, and address this problem again.

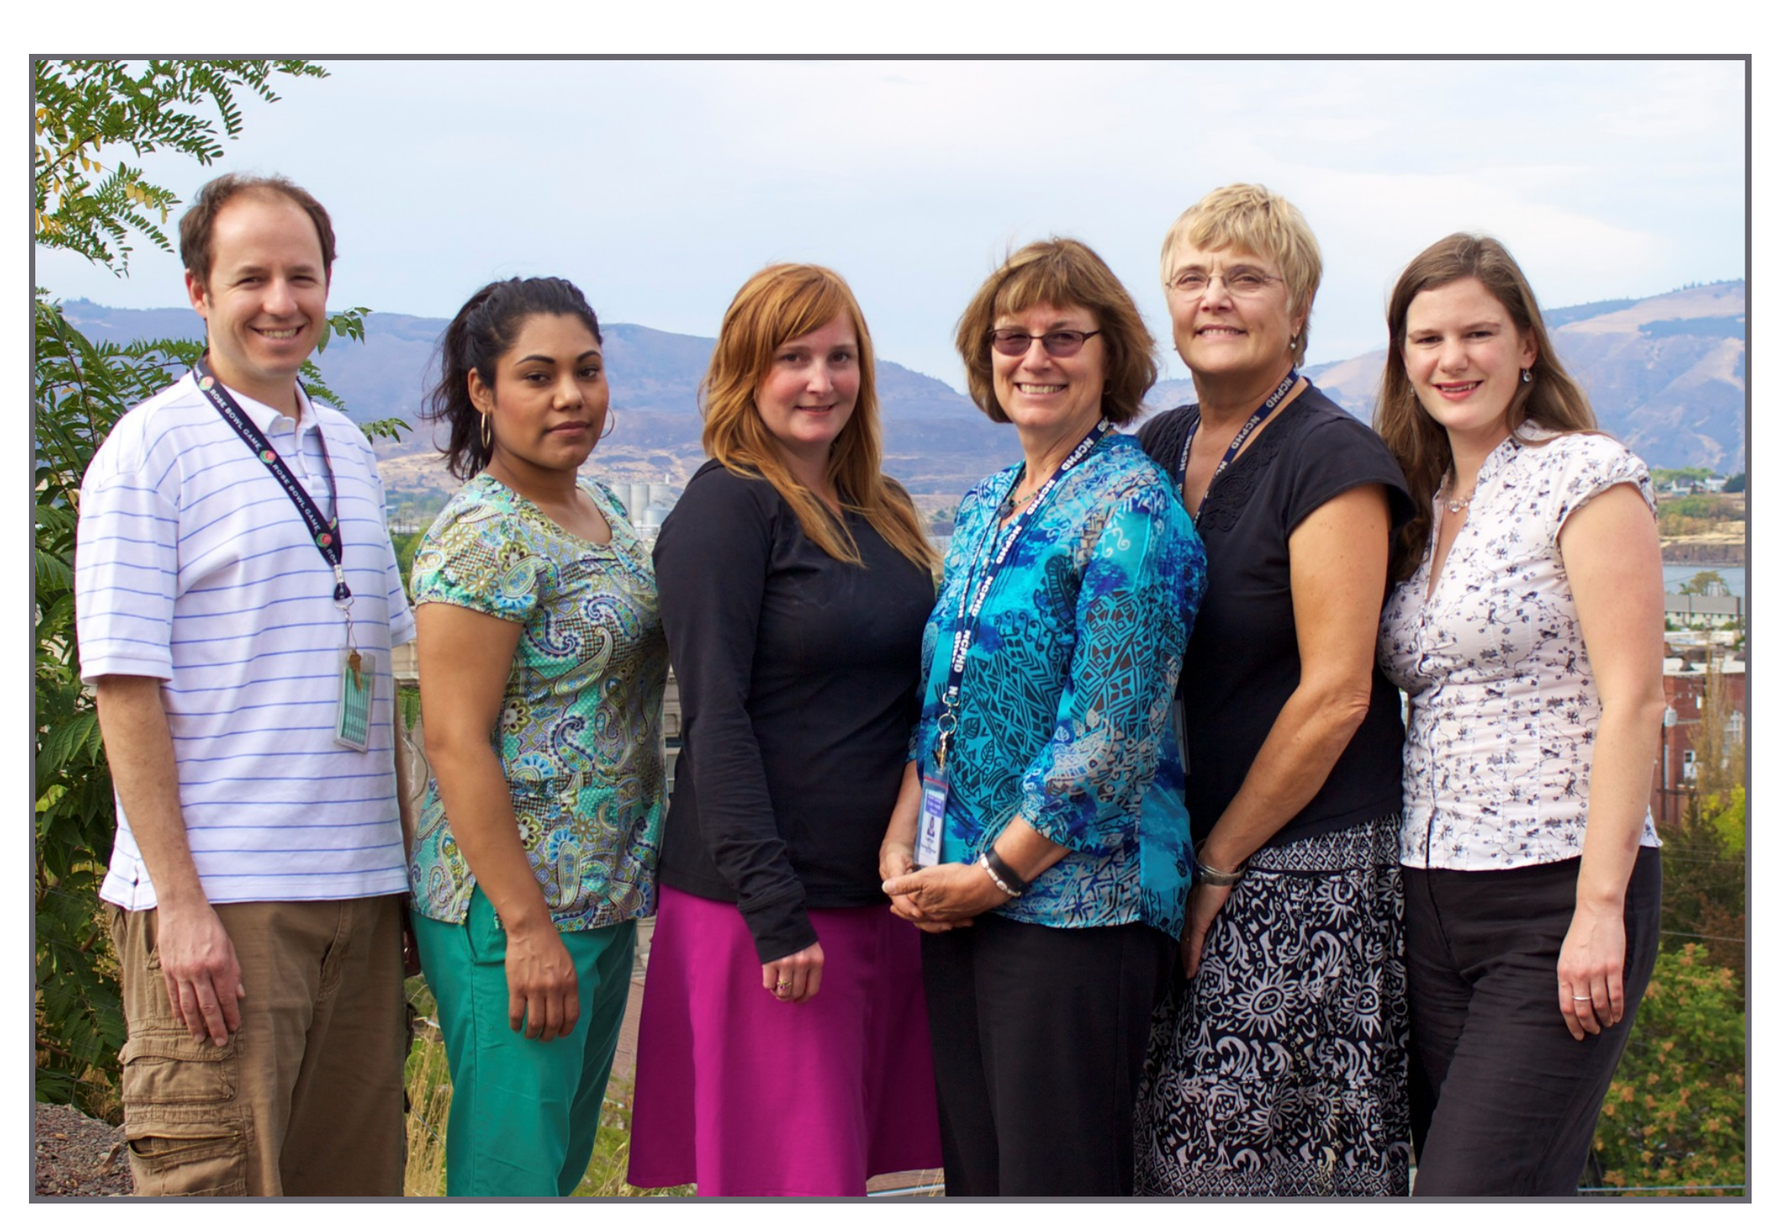

The QI Team (L to R)
Matthew Mercer – Front Office Specialist
Yary Ruiz – Family Planning Aide
Grace Anderson, RN - Clinic Nurse
Kathi Hall – Business Manager
Jane Palmer, RN – Nursing Supervisor
Faith Purvis, RN – Clinic Nurse
Tracy Willett, MD – Clinician
(in separate photo)

Our Providers:

Tracy Willett, MD from Sep 2011-Jan 2013

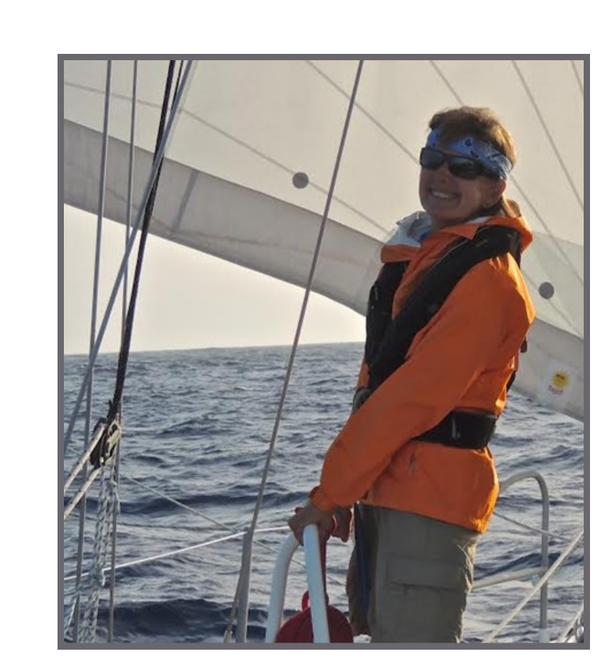

Lisa Nevara, FNP from Jan 2013-Current

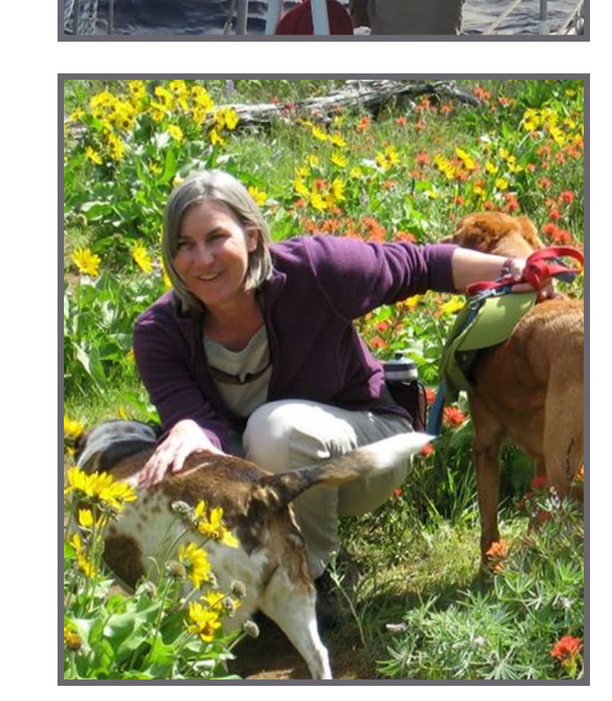

Reducing Clinic No-Show Rate QI Phase II: June 2014 - Current

Plan	Do	Check	Act
Improve the Reproductive Health Clinic Show Rate	1. Make reminder calls for both Monday and Tuesday appts on Friday; make additional reminder call for Tuesday appts on Monday. 2. Develop and implement system for text message appt reminders. 3. Restart follow up calls to assess barriers/ask why appt was missed. 4. Reassess days and demographics of no-shows 5. Continue using language emphasizing importance of keeping appt. 6. Continue clarifying what appt will entail/dispelling misconceptions.	- Prepare and compile data reports quarterly. - Assess new reminder call method: It appears effective. - F/u call results: limited success reaching clients, forgetfulness often given as reason by those who were reached. - Mondays found to have higher no-show rates than Tuesdays. **Findings at end of Sep 2014: Missed appointments reduced from 29% to 23%**	-Continue to develop text message reminder system. -Continue all strategies at this point. -Reassess quarterly and as needed. KEEP GOING